Primary Analysis of Phase 2 Results for Cemiplimab in Patients with Locally Advanced Basal Cell Carcinoma who Progress on or are Intolerant to Hedgehog Inhibitors

Alexander J. Stratigos,[1] Aleksandar Sekulic,[2] Ketty Peris,[3] Oliver Bechter,[4] Martin Kaatz,[5] Karl D. Lewis,[6] Nicole Basset-Seguin,[7] Anne Lynn S. Chang,[8] Stéphane Dalle,[9] Almudena Fernandez Orland,[10] Lisa Licitra,[11] Caroline Robert,[12] Claas Ulrich,[13] Axel Hauschild,[14] Michael R. Migden,[15] Reinhard Dummer,[16] Siyu Li,[17] Kosalai Mohan,[17] Ebony Coates,[17] Vladimir Jankovic,[17] Nathalie Fiaschi,[17] Emmanuel Okoye,[17] Ioannis Bassukas,[18] Carmen Loquai,[19] Vincenzo De Giorgi,[20] Zeynep Eroglu,[21] Ralf Gutzmer,[22] Jens Ulrich,[23] Susana Puig,[24] Frank Seebach,[17] Gavin Thurston,[17] Israel Lowy,[17] Timothy Bowler,[17] Matthew G. Fury[17]

[1]Andreas Sygros Hospital-University of Athens, Athens, Greece; [2]Arizona Mayo Clinic, Phoenix, AZ, USA; [3]Catholic University of the Sacred Heart and Fondazione Policlinico Universitario A. Gemelli-IRCCS, Rome, Italy; [4]Department of General Medical Oncology, University Hospitals, Leuven, Belgium; [5]SRH Wald-Klinikum Gera GmbH, Gera, Germany; [6]University of Colorado Hospital, Aurora, CO, USA; [7]Hopital Saint-Louis, Paris, France; [8]Department of Dermatology, Stanford University School of Medicine, Stanford, CA, USA; [9]Department of Dermatology, HCL Cancer Institute, Lyon Cancer Research Center, Lyon, France; [10]Hospital Universitario Virgen Macarena, Seville, Spain; [11]Fondazione IRCCS Istituto Nazionale dei Tumori and University of Milan, Milan, Italy; [12]Dermatology Unit, Gustave Roussy Cancer Center and Paris-Saclay University, Villejuif, France; [13]Charite-Universitiitsmedizin Berlin, Berlin, Germany; [14]Department of Dermatology, University of Kiel, Kiel, Germany; [15]The University of Texas MD Anderson Cancer Center, Houston, TX, USA; [16]University Hospital Zurich, Zurich, Switzerland; [17]Regeneron Pharmaceuticals, Inc., Tarrytown, NY, USA; [18]University of Ioánnina, Ioánnina, Greece; [19]Department of Dermatology, University Medical Center Mainz, Germany; [20]University of Florence, Florence, Italy; [21]Department of Cutaneous Oncology, Moffitt Cancer Center, Tampa, FL, USA; [22]Skin Cancer Center Hannover, Department of Dermatology and Allergy, Hannover Medical School, Hannover, Germany; [23]Skin Cancer Center, Department of Dermatology, Harz Clinics, Quedlinburg, Germany; [24]Hospital Clínic & Universitat de Barcelona and Centro de Investigación Biomédica en Red de Enfermedades Raras (CIBERER), Instituto de Salud Carlos III, Barcelona, Spain

Background

- Hedgehog inhibitors (HHIs), such as vismodegib and sonidegib, are approved for the treatment of patients with metastatic basal cell carcinoma (BCC) or locally advanced BCC (laBCC) who are not candidates for surgery or radiation.[1,2]
- However, for patients with laBCC, there is no standard regimen after first-line HHI therapy.[3]
- Cemiplimab is a high-affinity, highly potent, human programmed cell death (PD)-1 antibody, which has demonstrated anti-tumor activity in advanced solid tumors.[4–6]
- Cemiplimab is an established therapy approved for treatment of advanced cutaneous squamous cell carcinoma (CSCC) in patients who are not candidates for curative surgery or curative radiation.[7]
- Both BCC and CSCC are keratinocytic tumors with high mutational burden due to ultraviolet mutagenesis and are potentially amenable to immunotherapy.[3,8]
- We present the primary analysis of the laBCC cohort from the pivotal Phase 2 study of cemiplimab in the second-line (or greater) setting (NCT03132636).

Objectives

- The primary objective was to evaluate objective response rate (ORR) by independent central review (ICR).
- Key secondary endpoints: duration of response (DOR), progression-free survival (PFS), overall survival (OS), complete response by ICR, and safety and tolerability.
 - ORR included two responses confirmed after the data cut-off date of February 17, 2020.

Methods

- In this pivotal Phase 2 study, patients with laBCC received cemiplimab 350 mg every 3 weeks (Q3W) intravenously (IV) (for up to 93 weeks or until progression, unacceptable toxicity, withdrawal of consent, or confirmed complete response) (**Figure 1**).

Figure 1. Study design

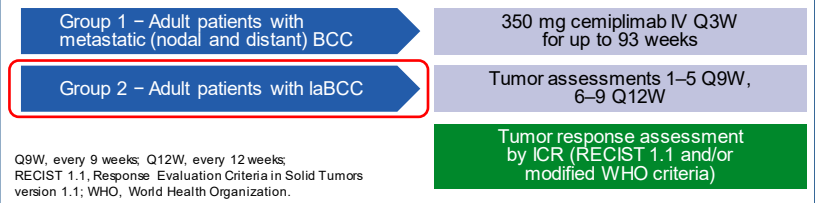
Group 1 − Adult patients with metastatic (nodal and distant) BCC
Group 2 − Adult patients with laBCC
350 mg cemiplimab IV Q3W for up to 93 weeks
Tumor assessments 1–5 Q9W, 6–9 Q12W
Tumor response assessment by ICR (RECIST 1.1 and/or modified WHO criteria)

Q9W, every 9 weeks; Q12W, every 12 weeks; RECIST 1.1, Response Evaluation Criteria in Solid Tumors version 1.1; WHO, World Health Organization.

- Key inclusion criteria:
 - Histologically confirmed diagnosis of invasive BCC
 - Prior progression or intolerance to HHI therapy or no better than stable disease after 9 months on HHI therapy
 - At least one measurable baseline lesion
 - Eastern Cooperative Oncology Group (ECOG) performance status of 0 or 1.
- Key exclusion criteria:
 - Ongoing or recent (within 5 years) autoimmune disease requiring systemic immunosuppression
 - Prior anti–PD-1 or anti–PD-ligand (L)1 therapy
 - Concurrent malignancy other than BCC and/or history of malignancy other than BCC within 3 years of date of first planned dose of cemiplimab, except for tumors with negligible risk of metastasis or death.

Results

Patient demographics and baseline characteristics

- As of February 17, 2020, 84 patients were enrolled; 66.7% were male; median age was 70 years (range: 42−89) (**Table 1**).
- The most common primary site of tumor location was head and neck (89.3%) (**Table 1**).
- Reasons for discontinuation of prior HHI therapy were progression of disease on HHI (71.4%), intolerant to prior HHI therapy (38.1%), and no better than stable disease after 9 months of HHI therapy (8.3%) (**Table 1**).
- Median follow-up was 15 months (range: 0.5−25).

Table 1. Patients demographics and baseline characteristics

	laBCC (N=84)
Median age, years (range)	70.0 (42–89)
Gender, % (n)	
Male	66.7 (56)
Female	33.3 (28)
ECOG performance status, % (n)	
0	60.7 (51)
1	39.3 (33)
Primary site of tumor, % (n)	
Head and neck	89.3 (75)
Extremity	2.4 (2)
Trunk	8.3 (7)
Reason for discontinuation of prior HHI, % (n)†	
Progression of disease on HHI	71.4 (60)
Intolerant to prior HHI therapy	38.1 (32)
Intolerant to vismodegib	38.1 (32)
Intolerant to sonidegib	4.8 (4)
No better than stable disease after 9 months on HHI therapy	8.3 (7)

†Sum is >84 because some patients had more than one reason for discontinuation.

Tumor response

- ORR per ICR was 31.0% (95% confidence interval [CI]: 21.3−42.0), including five complete responses and 21 partial responses (**Table 2**).
 - This includes two responses that emerged at the last tumor assessment prior to the data cut and were confirmed by ICR of tumor assessments after the data cut.

Table 2. Evaluation of response

n (%), unless otherwise stated	laBCC (N=84)
Objective response rate, % (95% CI)	31.0 (21.3−42.0)†
Complete response	5 (6.0)
Partial response	21 (25.0)
Stable disease	41 (48.8)
Progressive disease	9 (10.7)
Not evaluable‡	8 (9.5)

†Includes two patients whose partial responses were confirmed after the data cut-off date. ‡Of the eight patients who were not evaluable, four did not have any post-baseline tumor assessments, three patients were not considered to have evaluable lesions by either photographic or radiologic assessment methods, and one patient had a second target lesion not imaged after baseline.

- The estimated Kaplan–Meier probability of DOR was 90.9% (95% CI: 68.3−97.6) and 85.2% (95% CI: 60.5−95.0) at 6 and 12 months, respectively.
- Median DOR per ICR had not been reached at the time of data cut-off.
- Time to response and durability of responses were observed (**Figure 2**).
- Reductions of externally visible BCC lesions during cemiplimab treatment have been observed (**Figure 3**).
- Median estimated PFS for all patients was 19.3 months (95% CI: 8.6−not evaluable [NE]) (**Figure 4**).
- Median OS had not been reached at the time of data cut-off.
- The estimated 12-month probability of survival was 92.3% (95% CI: 83.6−96.5) (**Figure 4**).

Figure 2. DOR in responding patients

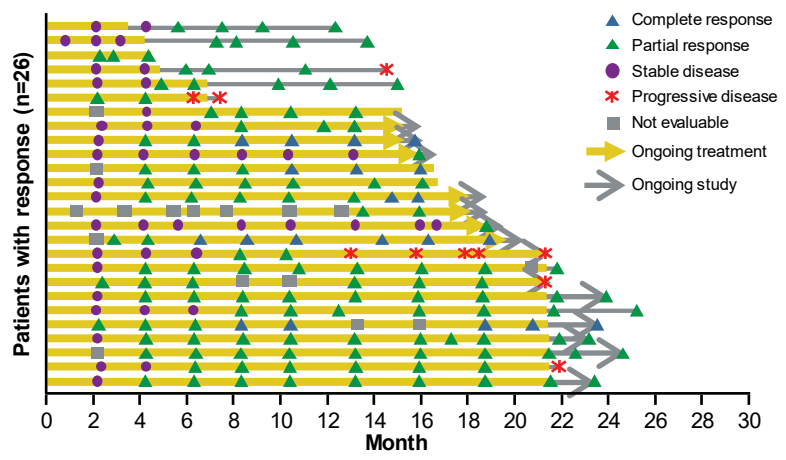
Patients with response (n=26)
Month
0 2 4 6 8 10 12 14 16 18 20 22 24 26 28 30
Complete response
Partial response
Stable disease
Progressive disease
Not evaluable
Ongoing treatment
Ongoing study

Figure 3. Reductions of visible BCC lesions while on cemiplimab treatment

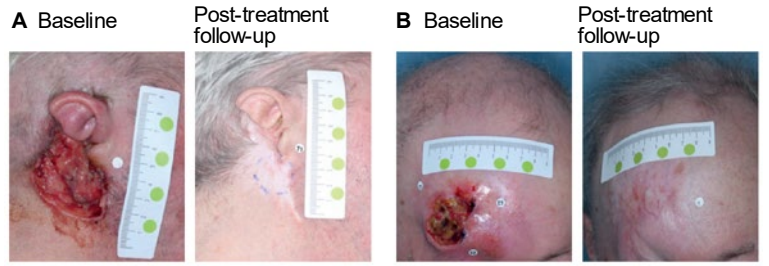
A Baseline
Post-treatment follow-up
B Baseline
Post-treatment follow-up

Panel A shows evidence of response from baseline (Study Day –24) to post-treatment follow-up (Study Day 726) in a 79-year-old man with progression of disease on prior vismodegib. Panel B shows response from baseline (Study Day –17) to post-treatment follow-up (Study Day 708) in a 66-year-old man who had received prior radiotherapy and prior vismodegib.

Figure 4. PFS and OS in responding patients

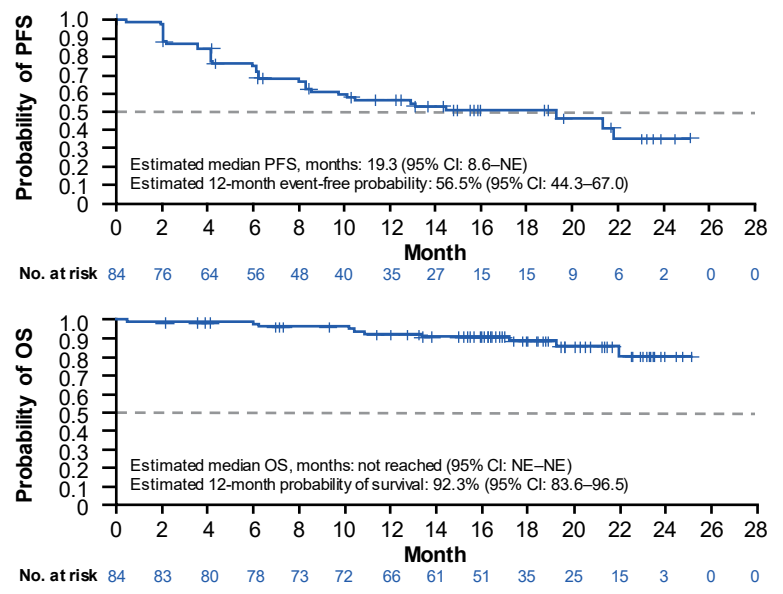
Probability of PFS
1.0 0.9 0.8 0.7 0.6 0.5 0.4 0.3 0.2 0.1 0
Estimated median PFS, months: 19.3 (95% CI: 8.6–NE)
Estimated 12-month event-free probability: 56.5% (95% CI: 44.3–67.0)
0 2 4 6 8 10 12 14 16 18 20 22 24 26 28
Month
No. at risk 84 76 64 56 48 40 35 27 15 15 9 6 2 0 0
Probability of OS
1.0 0.9 0.8 0.7 0.6 0.5 0.4 0.3 0.2 0.1 0
Estimated median OS, months: not reached (95% CI: NE–NE)
Estimated 12-month probability of survival: 92.3% (95% CI: 83.6–96.5)
0 2 4 6 8 10 12 14 16 18 20 22 24 26 28
Month
No. at risk 84 83 80 78 73 72 66 61 51 35 25 15 3 0 0

Tumor response by PD-L1 levels

- Baseline tumor samples were evaluable for PD-L1 in 50 of 84 patients (**Table 3**).
- ORR was 26% (95% CI: 13−43) in 35 patients with PD-L1 less than 1% and 27% (95% CI: 8−55) in 15 patients with PD-L1 ≥1% (**Table 3**).
- There was no evaluable PD-L1 in 34 patients (**Table 3**).
- Objective responses were observed in patients regardless of baseline PD-L1 levels (**Table 3**).

Table 3. Best objective tumor response rate by positive PD-L1 per ICR

n (%), unless otherwise stated	Evaluable PD-L1 (n=50)		No evaluable PD-L1 (n=34)
	PD-L1 <1% (n=35)	PD-L1 ≥1% (n=15)	(n=34)
Objective response rate, % (95% CI)	26 (13−43)	27 (8−55)	38 (22−56)
Complete response	2 (6)	2 (13)	1 (3)
Partial response	7 (20)	2 (13)	12 (35)
Stable disease	18 (51)	9 (60)	14 (41)
Progressive disease	5 (14)	1 (7)	3 (9)
Not evaluable	3 (9)	1 (7)	4 (12)
Disease control rate, % (95% CI)†	77 (60−90)	87 (60−98)	79 (62−91)
Durable disease control rate, % (95% CI)‡	51 (34−69)	53 (27−79)	71 (53−85)

†Defined as the proportion of patients with complete response, partial response, stable disease, or non-partial response/non-progressive disease at the first evaluable tumor assessment, scheduled to occur at week 9 (defined as 56 days to account for visit windows in the protocol). ‡Defined as the proportion of patients with complete response, partial response, stable disease, or non-partial response/non-progressive disease for at least 27 weeks without progressive disease (defined as 182 days to account for visit windows in the protocol).

Safety

- The most common treatment-related adverse events (TRAEs), by preferred terms, included fatigue (n=21; 25%), pruritus (n=12; 14%), and asthenia (n=12; 14%).
- The most common Grade ≥3 TRAEs were colitis (n=4), fatigue, and adrenal insufficiency (n=2 each).
- Immune-related adverse events (irAEs, per sponsor identification method) occurred in 21 (25%) patients and were Grade 3 in eight (10%) patients. No Grade 4 or Grade 5 irAEs occurred.
- The most common irAEs (any grade) were hypothyroidism and colitis, in eight (10%) and five (6%) patients, respectively.
- Grade 3 irAEs in >1 patient were colitis (n=3) and adrenal insufficiency (n=2).
- Adverse events of Grade 3 or greater, regardless of attribution, occurred in 51% of patients (**Table 4**).
- Seventeen percent of patients discontinued treatment due to TEAEs (**Table 4**).
- There were no treatment-related deaths (**Table 4**).
- The overall safety profile is generally consistent with previously reported experience with cemiplimab.

Table 4. TEAEs regardless of attribution

n (%)	laBCC (N=84)	
	Any grade	Grade ≥3
Any	82 (98)	43 (51)
Serious	29 (35)	22 (26)
Led to discontinuation	14 (17)	7 (8)
Associated with an outcome of death†	3 (4)	3 (4)
Occurred in ≥10% patients (any grade) or Grade 3 in ≥5% patients		
Fatigue	25 (30)	3 (4)
Diarrhea	20 (24)	0
Pruritus	18 (21)	0
Asthenia	17 (20)	1 (1)
Anemia	13 (16)	1 (1)
Decreased appetite	13 (16)	1 (1)
Headache	12 (14)	1 (1)
Nausea	12 (14)	1 (1)
Urinary tract infection	12 (14)	3 (4)
Arthralgia	11 (13)	0
Dyspnea	10 (12)	0
Blood creatinine increased	8 (10)	0
Dizziness	8 (10)	0
Cough	8 (10)	0
Hypothyroidism	8 (10)	0
Tumor hemorrhage	8 (10)	0
Hypertension	7 (8)	4 (5)

†Not considered treatment-related. TEAE, treatment-emergent adverse event.

Summary and Conclusion

- Cemiplimab is the first systemic therapy to show clinical benefit in patients with laBCC after HHI therapy with a 31.0% ORR per ICR. Among responders, the estimated 12-month DOR was 85.2%.
- The safety profile is generally consistent with other PD-1 antibodies and with previous reports of cemiplimab in other tumor types.
- Baseline PD-L1 expression is not associated with clinical activity.
- Combined with interim data from the metastatic BCC cohort,[9] these results confirm that cemiplimab has substantial activity in advanced BCC tumors.

References

1. Sun Pharmaceuticals Industries, Inc. ODOMZO (®sonidegib) prescribing information. Available from: https://www.accessdata.fda.gov/drugsatfda_docs/label/2017/205266s004lbl.pdf. Revised September 2017. Accessed October 12, 2020.
2. Genentech, Inc. ERIVEDGE™ (vismodegib) prescribing information. Available from: https://www.accessdata.fda.gov/drugsatfda_docs/label/2012/203388lbl.pdf. Revised January 2012. Accessed October 12, 2020.
3. Goodman AM et al. *Oncoimmunology* 2018;7:e1404217.
4. Burova E et al. *Mol Cancer Ther.* 2017;16:861–870.
5. Migden MR et al. *N Engl J Med.* 2018;379: 341–351.
6. Rischin D et al. *J Immunother Cancer.* 2020;8:e000775.
7. Regeneron Pharmaceuticals, Inc. and Sanofi-Aventis US LLC. LIBTAYO® [cemiplimab-rwlc] injection full US prescribing information. Available from: https://www.accessdata.fda.gov/drugsatfda_docs/label/2018/761097s000lbl.pdf. Accessed July 13, 2020.
8. Inman GJ et al. *Nat Commun.* 2018;9:3667.
9. Lewis KD et al. Poster presented at the SITC Virtual Congress 2020. November 9–14, 2020: Poster 428.

Acknowledgments

The authors would like to thank the patients, their families, investigators, and all investigational site members involved in this study. The study was funded by Regeneron Pharmaceuticals, Inc. and Sanofi. Editorial writing support was provided by Jenna Lee of Prime, Knutsford, UK, funded by Regeneron Pharmaceuticals, Inc. and Sanofi.

Disclosures

Alexander J. Stratigos reports advisory board or steering committee roles with Janssen CILAG, Regeneron Pharmaceuticals, Inc., Roche, and Sanofi, and research support from Abbvie, BMS, Genesis Pharma, and Novartis. **Aleksandar Sekulic** reports advisory roles with Regeneron Pharmaceuticals, Inc. and Roche. **Ketty Peris** reports advisory board roles with Abbvie, LEO Pharma, Janssen, Almirall, Lilly, Galderma, Novartis, Pierre Fabre, Sun Pharma, and Sanofi outside the submitted work. **Oliver Bechter** has advisory board roles with Novartis, BMS, Merck Sharp & Dhome, Pierre Fabre, and Ultimovacs. **Martin Kaatz, Almudena Fernandez Orland, Ioannis Bassukas,** and **Vincenzo De Giorgi** declare no conflict of interest. **Karl D. Lewis** reports grants from the University of Colorado and grants and personal fees from Regeneron Pharmaceuticals, Inc. **Nicole Basset-Seguin** reports honoraria from Galderma, Leo, Pierre Fabre, Novartis, and Roche, consulting fees from Galderma, Leo, Pierre Fabre, Novartis, Roche, and Sun Pharmaceuticals, Inc. patents, royalties, or other intellectual property from Genentech/F. Hoffmann-La Roche, Ltd., and travel, accommodations, or expenses from Galderma, Leo, and Roche. **Anne Lynn S. Chang** reports advisory roles with Regeneron Pharmaceuticals, Inc. and Merck, and research funding from Regeneron Pharmaceuticals, Inc., Merck, Novartis, and Galderma. **Stéphane Dalle** reports that their spouse is an employee of Sanofi. **Lisa Licitra** reports institutional grants and personal fees from AstraZeneca, BMS, Boehringer Ingelheim, Debiopharm International SA, Eisai, Novartis, and Roche, institutional grants from Celgene International, Exelixis Inc, Hoffmann-La Roche Ltd, IRX Therapeutics Inc.,Medpace Inc., Merck-Serono, and Pfizer, and personal fees from Sobi, Ipsen, Incyte Biosciences Italy SRL, Doxa Pharma, Amgen, Nanobiotics SA, GSK, AccMed, Medical Science Fundation G. Lorenzini, Associazione Sinapsi, Think 2 IT, Aiom Servizi, Prime Oncology, WMA Congress Education, Fasi, DueCi Promotion SRL, MI&T, Net Congress & Education, PRMA Consulting, Kura Oncology, Health & Life SRL, and Immuno-Oncology Hub. **Caroline Robert** reports grants and personal fees advisory board roles with Bristol-Myers Squibb, Pierre Fabre, Novartis, Amgen, Merck, Roche, Merck Sharp & Dhome, Sanofi, Biothera, and Ultimovacs. **Claas Ulrich** reports advisory board and speaker roles for Roche, Sanofi, Regeneron Pharmaceuticals, Inc., and Sun Pharma. **Axel Hauschild** reports institutional funding and personal fees from Amgen, BMS, MerckSerono, Merck Sharp & Dhome/Merck, Philogen, Pierre Fabre, Provectus, Regeneron Pharmaceuticals, Inc., Roche, Sanofi-Genzyme, Novartis, and consultancy fees from OncoSec, Almirall Hermal, and Sun Pharma. **Michael R. Migden** reports advisory roles with and travel fees from Regeneron Pharmaceuticals, Inc. and Sun Pharmaceuticals, advisory role with Rakuten Medical, and research funding from Regeneron Pharmaceuticals, Inc. and Pelle Pharm. **Reinhard Dummer** reports consultant or advisory roles with Novartis, Merck Sharp & Dhome, BMS, Roche, Amgen, Takeda, Pierre Fabre, Sun Pharma, Sanofi, Catalym, Second Genome, Regeneron Pharmaceuticals, Inc.,and Alligator outside the submitted work. **Siyu Li, Kosalai Mohan, Ebony Coates, Vladimir Jankovic, Nathalie Fiaschi, Emmanuel Okoye, Frank Seebach, Gavin Thurston,** and **Timothy Bowler** are employees and shareholders of Regeneron Pharmaceuticals, Inc. **Carmen Loquai** reports personal fees from Roche, Sun Pharma, BMS, MSD, Merck, Novartis, Pierre Fabre, Kyowa Kirin, Almiral Hermal, and Biontech. **Zeynep Eroglu** reports advisory board fees from Regeneron Pharmaceuticals, Inc., Novartis, Array, Genentech, and SunPharma, and grant from Novartis. **Ralf Gutzmer** reports documentation fees to institution from Regeneron Pharmaceuticals, Inc.; personal fees and non-financial support from Amgen, BMS, Roche Pharma, Merck Serono, Pierre Fabre, Sanofi, and Regeneron Pharmaceuticals, Inc.; grants, personal fees and non-financial support from Novartis; grants and personal fees from Pfizer; grants from Johnson & Johnson; and personal fees from Merck Sharp Dohme, Almirall Hermal, Sun Pharma, 4SC, and Bayer. **Jens Ulrich** reports grants and personal fees from Sanofi, BMS, Novartis, and MSD; personal fees from Roche, medac, and Sun Pharma. **Susana Puig** reports personal fees and non-financial support from Sanofi; other (clinical trials) from Regeneron Pharmaceuticals, Inc.; grants from Avene; non-financial support from Abbie; grants, personal fees and non-financial support from ISDIN, La Roche-Posay, and Roche; grants and personal fees from Sun Pharma; non-financial support from Lilly and MSD; personal fees, non-financial support and other from Pfizer; grants, personal fees and other from Leo Pharma and Almirall; personal fees and other from Ojer Pharma; personal fees from Pierre Fabre; non-financial support from MAVIG, FotoFinder, and 3Gen; personal fees, non-financial support and other from Novartis, grants and personal fees from Amgen. **Israel Lowy** and **Matthew G. Fury** are employees of, have patents pending, and are shareholders of Regeneron Pharmaceuticals, Inc.

This poster is available to download here:

Copyright © 2021 A.J. Stratigos et al. For questions, please contact Alex Stratigos <alstrat2@gmail.com>

Presented at the Joint Virtual Scientific Meeting 2021 ANZHNCS and NZAPS, August 5–8, 2021 (encore of ESMO 2020 late-breaking mini oral presentation).